Prevention of *Pneumocystis* Pneumonia by Ibrexafungerp in a Murine Prophylaxis Model.

Stephen Barat[1], Katyna Borroto-Esoda[1], Alan Ashbaugh [2,3], Melanie Cushion [2,3]
[1]SCYNEXIS, Inc., [2]University of Cincinnati Medical Center, [3]Veteran Affairs Medical Center and Cincinnati Education and Research for Veterans Foundation , Cincinnati, OH

Background: *Pneumocystis* pneumonia (PCP) is an opportunistic fungal infection that affects immunocompromised patients. Ibrexafungerp (IBX) is an oral and intravenous antifungal from a novel class of glucan synthase inhibitors, triterpenoids, and has shown activity against *Candida*, *Aspergillus*, and PCP in a murine therapy model. We evaluated the ability of IBX to prevent PCP in a prophylaxis model of murine PCP.

Materials/methods: Experiment 1: Balb/c mice (10 mice/group) were infected by intranasal inoculation with *Pneumocystis murina*, immune-suppressed with dexamethasone in acidified drinking water and treated with 30-, 15- and 7.5 mg/kg, IBX/BID. Control groups treatment included TMP-SMX (50/250 mg/kg QD) and vehicle. After 6 weeks, mice were sacrificed, and prevention was determined by organism burdens (asci and total nuclei). **Experiment 2**: Balb/c mice were immune-suppressed and infected as in Exp. 1. Treatment groups included: 1) 30 mg/kg BID x 6wk; 2) 30 mg/kg/BID x 6wk followed by cessation of treatment with IBX but with immune-suppression for 3 additional weeks; 3) 15 mg/kg BID 1 week prior and 6 wks after infection and immune suppression; 4) 15mg/kg BID for 8 wks; 5) 15 mg/kg BID for 6 wks then IBX was discontinued but with immune suppression; 6) untreated, vehicle control.

Results: Experiment 1: No *P. murina* nuclei or asci were observed after 6 weeks of treatment at a dose of 30 mg/kg/BID in the prophylaxis mouse model of PCP, similar to positive control, TMP/SMX. Some nuclei and asci were observed in the lower dose IBX groups. **Experiment 2:** To investigate whether any *P. murina* remained after different regimens of prophylaxis, treatment of IBX was withdrawn at both doses for an additional 3 wks of immune suppression to provoke the growth of any remaining fungi. Group 1 showed reduction in total nuclei and asci to undetectable. Group 2 did not result in any recrudescence of infection. Group 3 and 4 showed similar reduction in organism burden. Group 5 was similar to untreated control.

Conclusions: These results demonstrate that 30 mg/kg BID IBX prevented PCP in a murine model. We suggest that IBX could be a viable option for preventing PCP in immunocompromised patients.