A Novel Protocol Design to Study the Efficacy and Safety of Oral Ibrexafungerp as Step-Down Therapy following Intravenous (IV) Echinocandin for the Treatment of Invasive Candidiasis (MARIO): Developing A Paradigm Shift to IV and Oral Anti-Cell Wall Therapy

P2055

TJ Walsh [1], L Ostrosky-Zeichner[2], OA Cornely[3], J Vazquez[4], BJ Kullberg[5], A Spec[6], NE Azie[7], DA Angulo[7], and PG Pappas[8]

[1]Center for Innovative Therapeutics and Diagnostics, Richmond, VA, USA, [2]University of Texas, Houston, USA, [3]University of Cologne, Cologne, Germany, [4]Augusta University, Augusta, GA, USA, [5]Raboud University, Nijmegen, Netherlands, [6]Washington University at St. Louis, MO, USA, [7]SCYNEXIS, Inc., Jersey, City, NJ, USA, [8]University of Alabama, Birmingham, AL USA

33rd ECCMID EUROPEAN CONGRESS OF CLINICAL MICROBIOLOGY AND INFECTIOUS DISEASES

BACKGROUND

There are limited oral therapies available to treat invasive fungal diseases. Current treatment guidelines for invasive candidiasis (IC) and candidemia (C) recommend use of either (1) an intravenous (IV) echinocandin followed by oral fluconazole for susceptible species, or (2) continuing the echinocandin, or (3) changing to oral voriconazole/high-dose fluconazole for fluconazole non-susceptible species (Figure 1). Currently there are limited oral options for patients with triazole-resistant *Candida* species. Ibrexafungerp (IBX) is a novel triterpenoid antifungal agent with activity against fluconazole- and echinocandin-susceptible and -resistant strains, being developed as the first oral glucan synthase inhibitor for the treatment of IC/C. An ongoing study explores the efficacy and safety of oral IBX for the step-down treatment of invasive candidiasis and candidemia following an IV echinocandin (ClinicalTrials.gov Identifier NCT05178862). We present herein an innovative trial-design that may change the paradigm to echinocandin followed by IBX for the treatment of subjects with invasive candidiasis and candidemia.

Figure 1: Treatment options for Invasive Candidiasis by *Candida* spp.

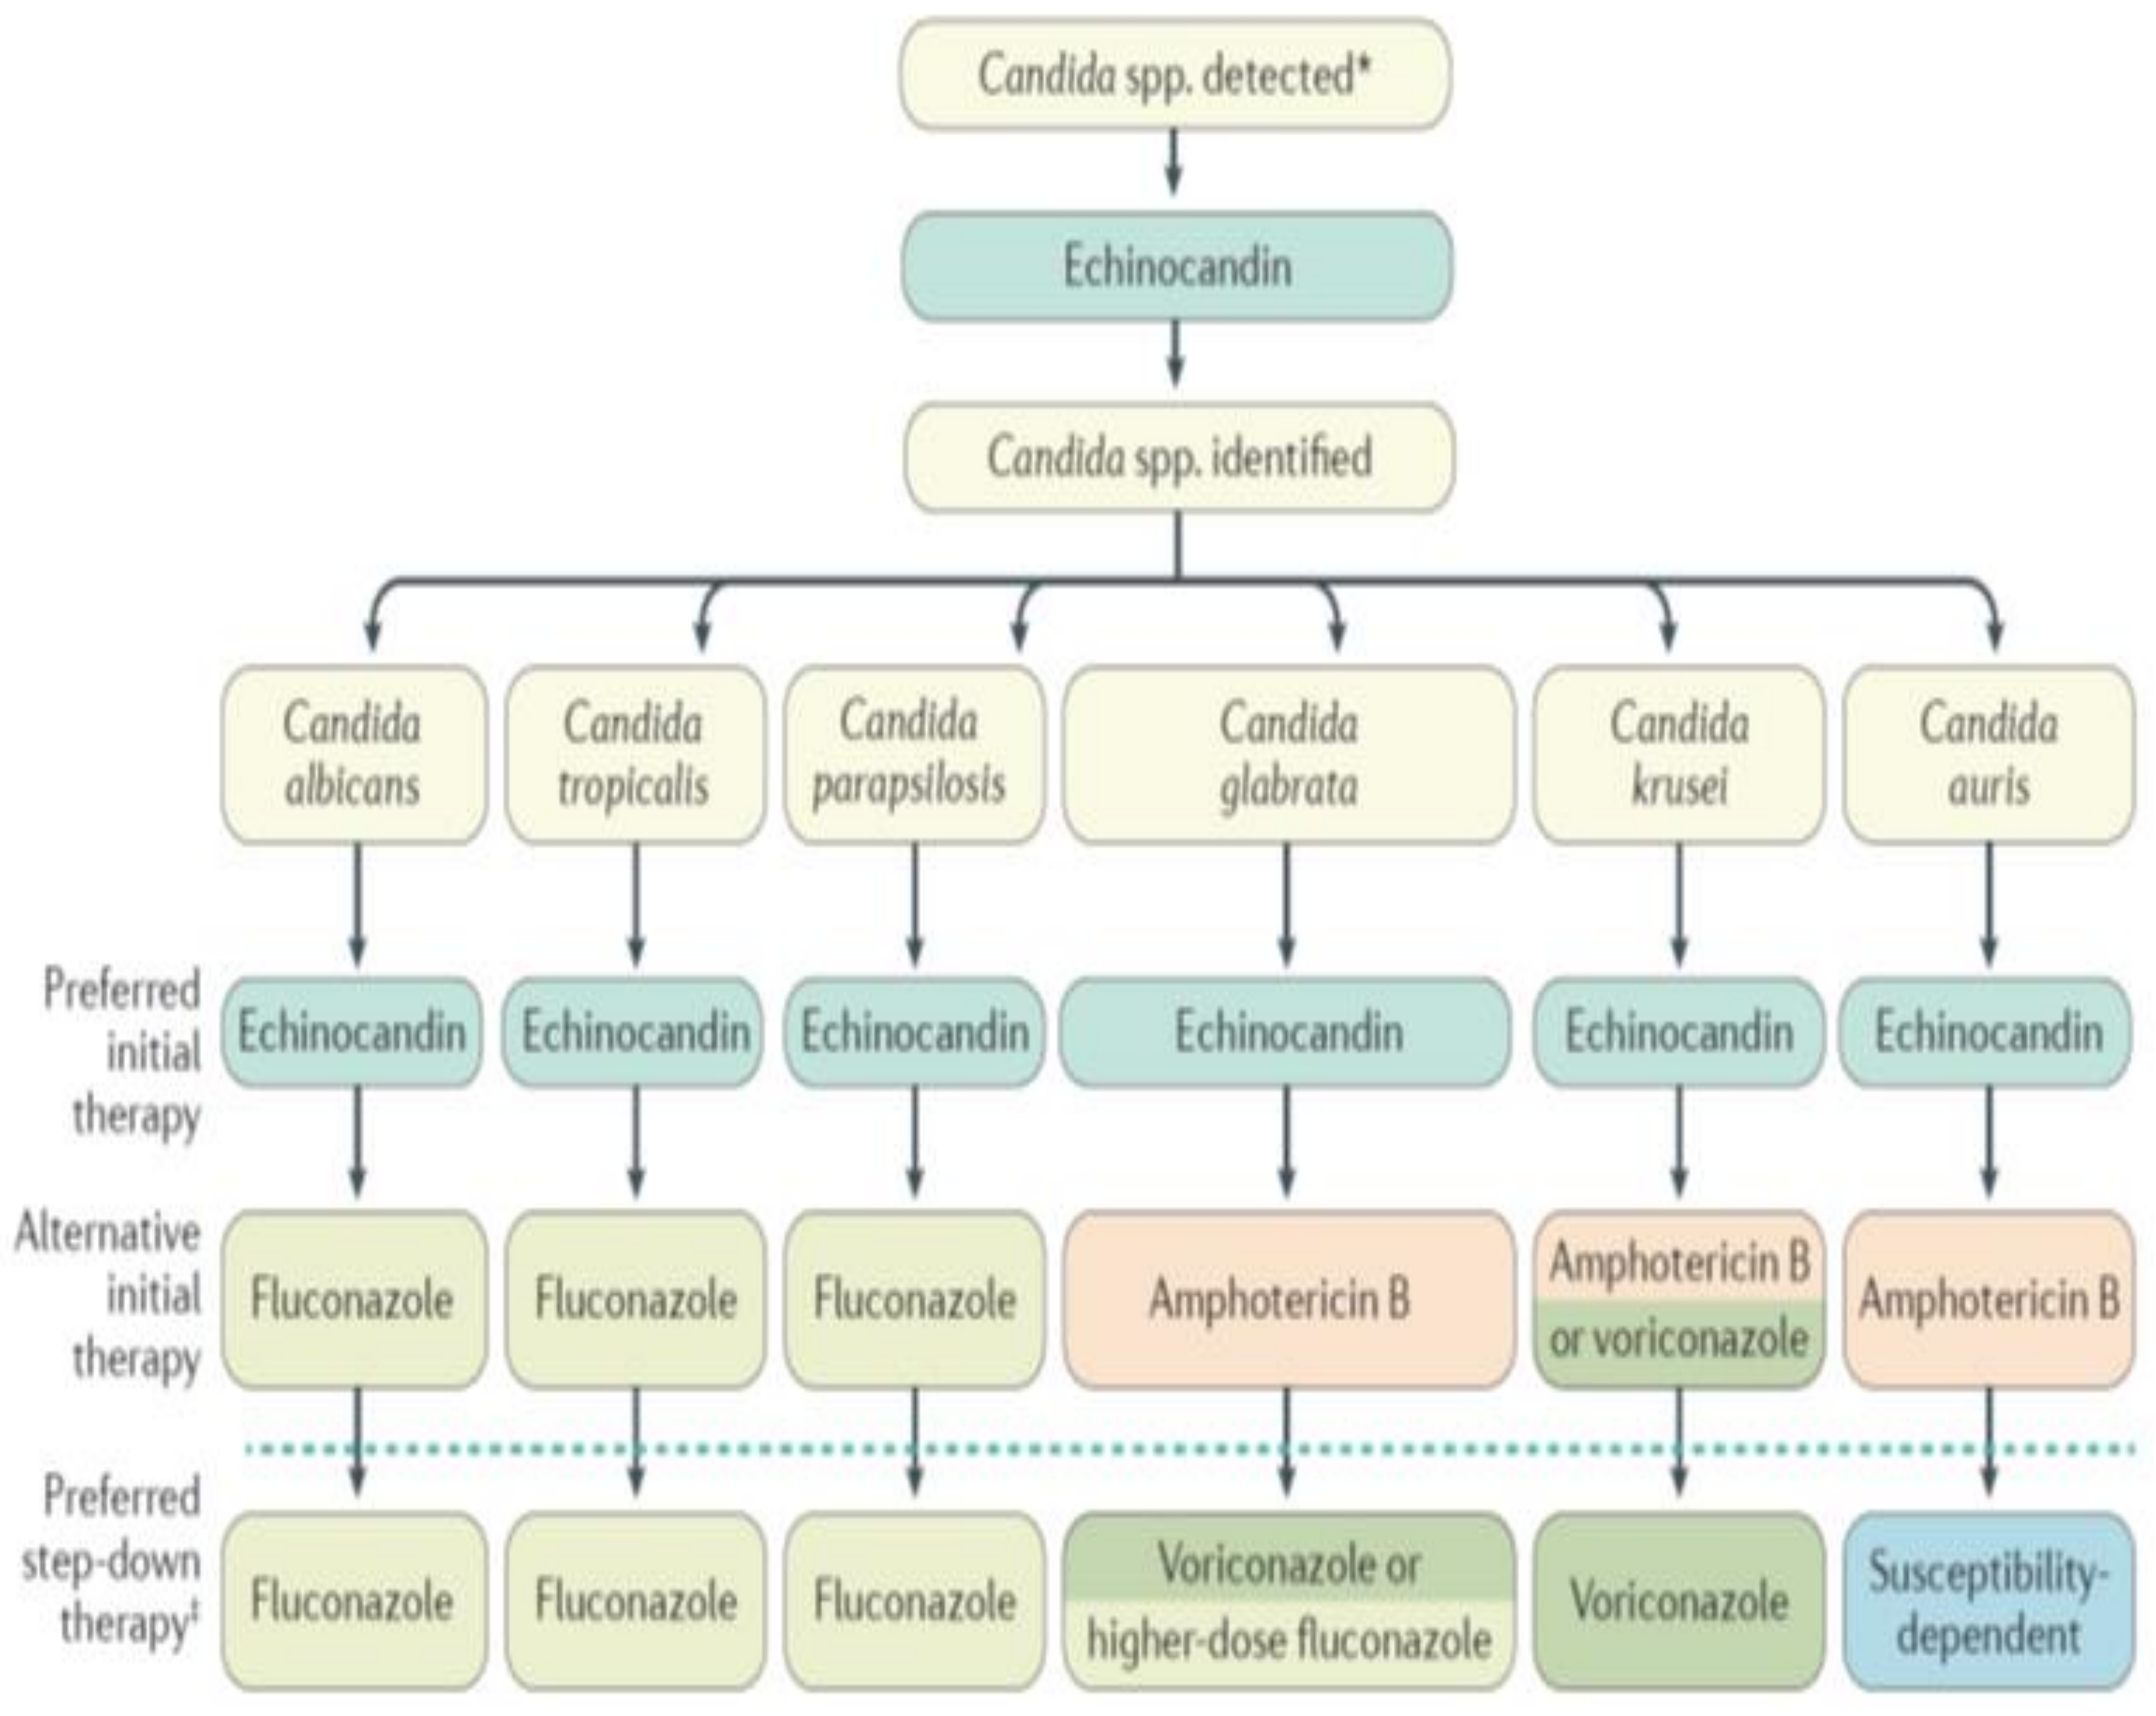

Candida spp. detected*
Echinocandin
Candida spp. identified
Candida albicans
Candida tropicalis
Candida parapsilosis
Candida glabrata
Candida krusei
Candida auris
Preferred initial therapy
Echinocandin
Alternative initial therapy
Fluconazole
Amphotericin B
Amphotericin B or voriconazole
Preferred step-down therapy‡
Voriconazole or higher-dose fluconazole
Voriconazole
Susceptibility-dependent

Adapted from Pappas et al. Nature Reviews doi:10.1038/nrdp.2018.26

STUDY DESIGN

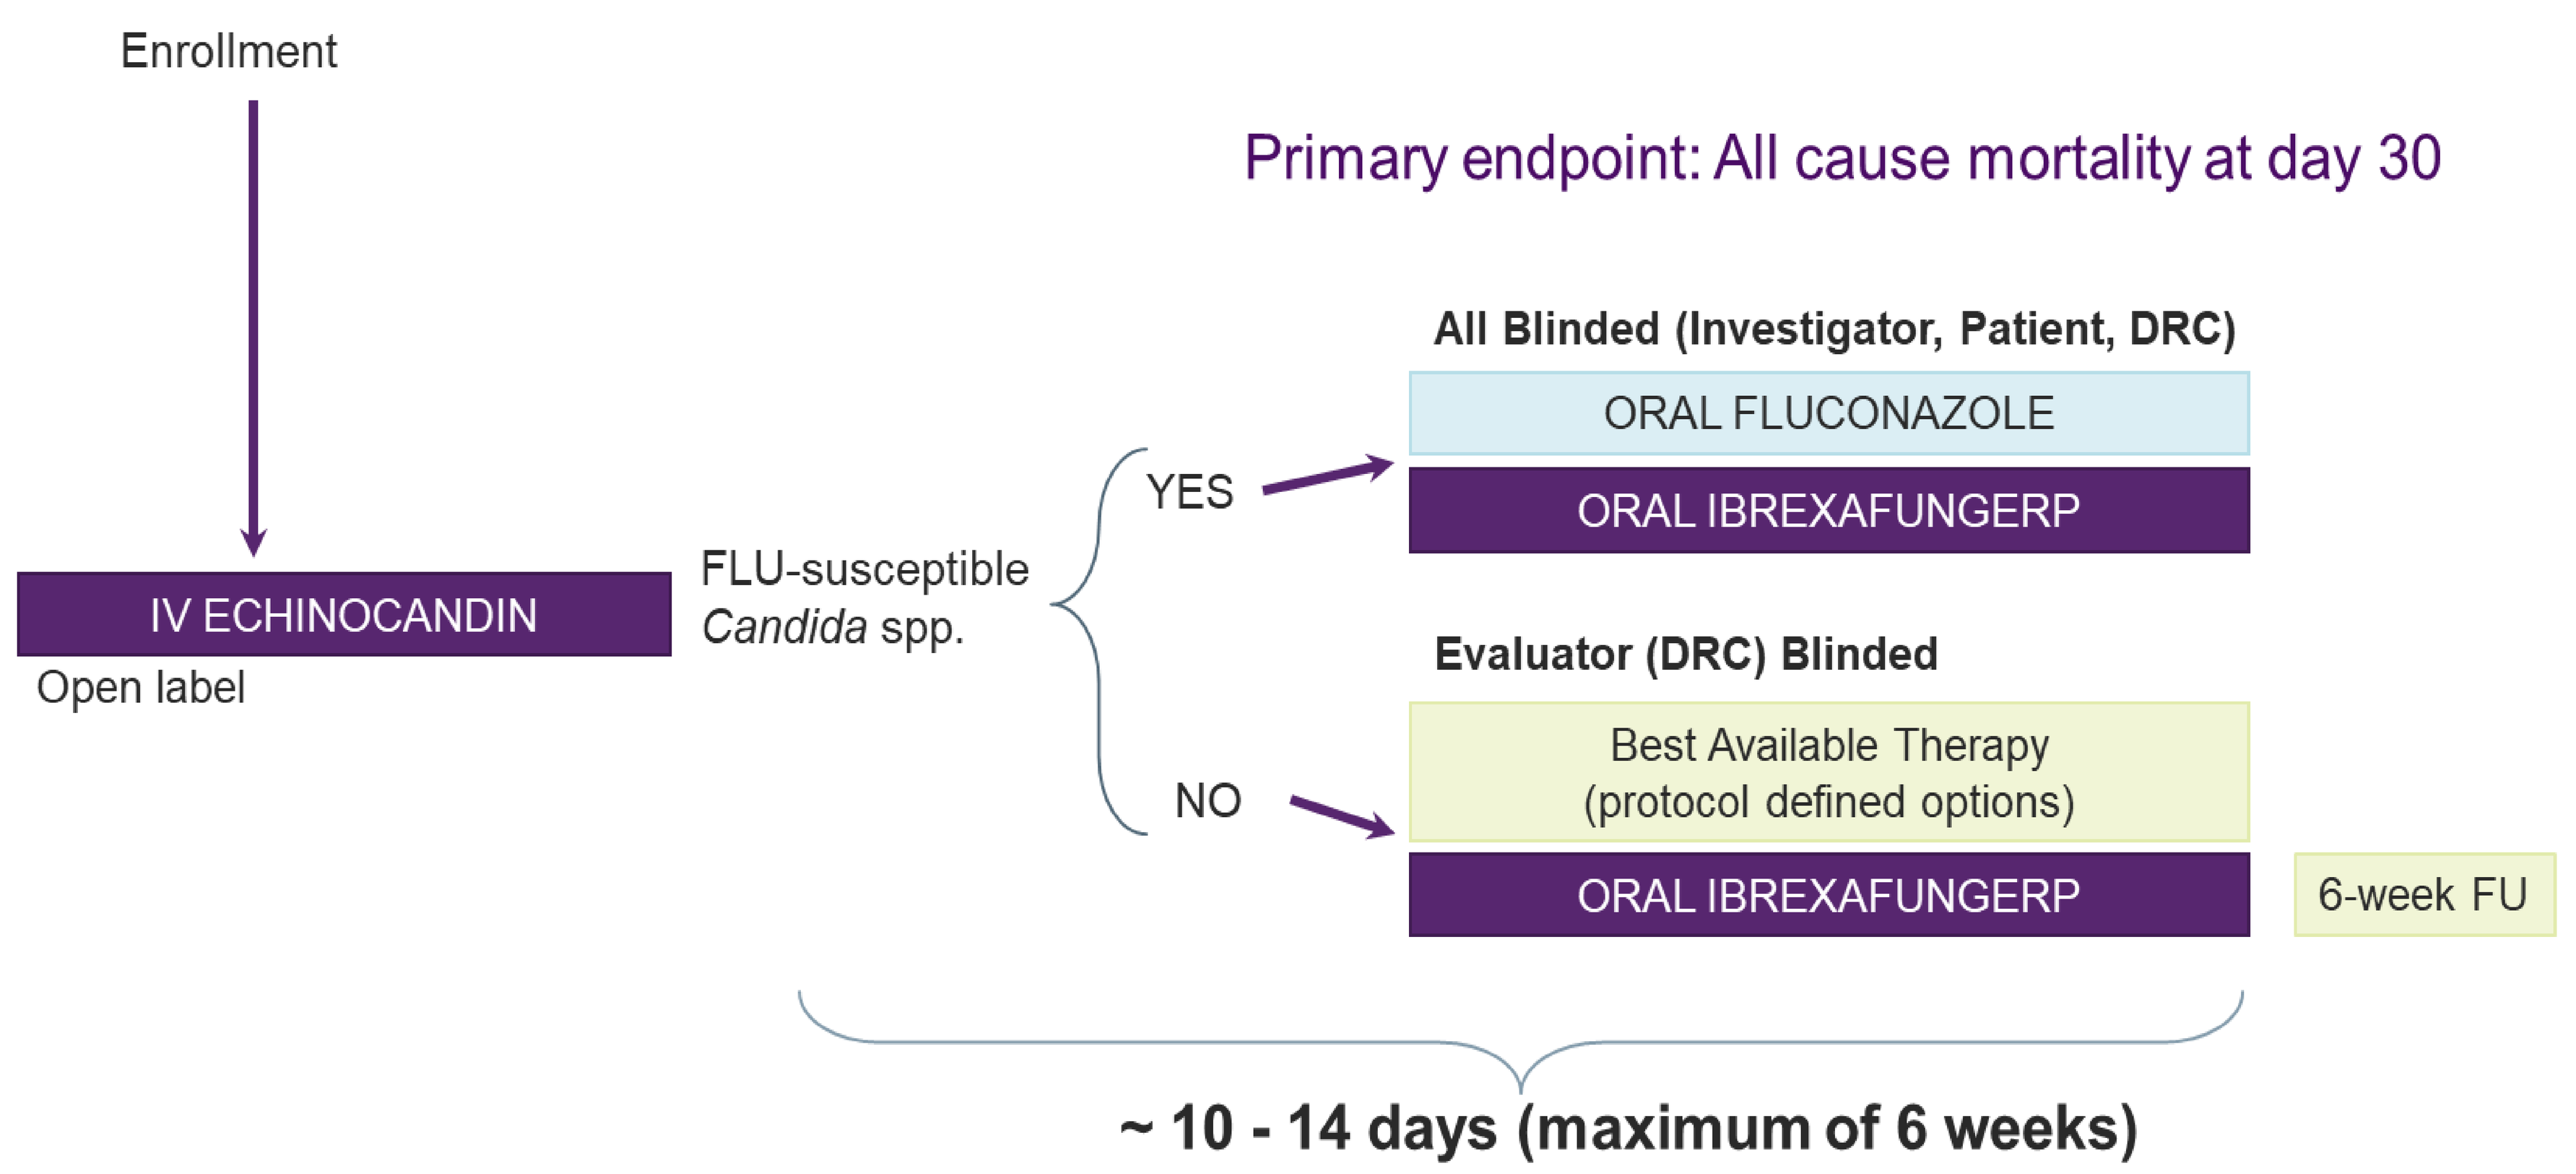

Enrollment
Primary endpoint: All cause mortality at day 30
All Blinded (Investigator, Patient, DRC)
ORAL FLUCONAZOLE
YES
ORAL IBREXAFUNGERP
IV ECHINOCANDIN
Open label
FLU-susceptible Candida spp.
Evaluator (DRC) Blinded
Best Available Therapy (protocol defined options)
NO
ORAL IBREXAFUNGERP
6-week FU
~ 10 - 14 days (maximum of 6 weeks)

Figure 2: MARIO Study Design Schematic

- This is a Phase 3, multicenter, randomized, double-blind, double-dummy, active-controlled, comparative study to evaluate the efficacy, safety and tolerability of oral IBX compared to oral fluconazole (FLU) step-down treatment following IV echinocandin in the treatment of adult subjects (≥18 years old) with invasive candidiasis and/or candidemia. The pharmacokinetics of IBX will also be described.
- Eligible subjects will receive initial treatment with IV echinocandin. Treatment will then be switched to double-blind, double-dummy oral therapy (either IBX or FLU) as soon as step-down criteria are met. Subjects with FLU non-susceptible isolates will receive open-label oral IBX or Best Available Therapy (BAT) (Figure 2).

Key Inclusion Criteria

- Male or female adult ≥ 18 years of age on the day the study informed consent is signed.
- Subject has a diagnosis of candidemia and/or invasive candidiasis, defined as evidence of *Candida* spp. in either a bloodstream or tissue culture from a normally sterile site collected ≤ 4 days (within 96 hours) prior to initiation of IV echinocandin accompanied by any related clinical sign and/or symptom (e.g., fever [on one occasion > 38°C], hypotension, or local signs of inflammation).
- Subject is not pregnant or lactating and not planning to become pregnant

Key Exclusion Criteria

Subject has any of the following forms of invasive candidiasis at Screening:

- Septic arthritis in a prosthetic joint (septic arthritis in a native joint is allowed), osteomyelitis, endocarditis or myocarditis, meningitis, endophthalmitis, or any central nervous system infection, chronic disseminated candidiasis
- Patients with concurrent invasive fungal infection other than *Candida* spp.
- Patients who failed a previous antifungal therapy for the same infection
- Subject has an inappropriately controlled fungal disease source (e.g., indwelling vascular catheter or device that cannot be removed or an abscess that cannot be drained) that is likely to be the source of the IC or C

Study Highlights

- First study to compare efficacy of oral step-down therapies from IV echinocandin. The study compares oral IBX vs Standard-of Care (oral azole or continued IV therapy) for treatment of IC or C
- Treatment guidelines recommend a glucan synthase inhibitor (IV echinocandin) as preferred first-line therapy. The study is the first to offer opportunity to continue therapy with an **Oral** glucan synthase inhibitor (ibrexafungerp), for the treatment of IC or C.
- Patients can be identified and enrolled even after initiation of echinocandin therapy.

Dosing

All patients are started on an IV echinocandin followed by,

- Oral IBX 750 mg BID Loading dose on Days 1 and 2, followed by oral IBX 750 mg QD;
- Or oral FLU 800 mg QD loading dose on Day 1 followed by oral FLU 400 mg QD (See Figure 1)

Symptoms of IC/Candidemia- include, <u>but are not limited</u> to, the following:

- Fever defined as oral temperature equivalent to ≥ 38.3°C (≥ 101°F) on one occasion or > 37.8°C (> 100°F) on two measurements at least 4 hours apart
- Clinically significant hypothermia < 36°C (< 96.8°F)
- Hypotension (systolic blood pressure of < 90 mmHg or > 30 mmHg decrease below normal baseline)
- Signs of inflammation at the *Candida* infected site

Criteria for Oral Switch

IV Echinocandin treatment ideally for 5 days until criteria for oral switch are met:

- Subjects are clinically stable
- Able to tolerate oral medication
- Hemodynamically stable
- No post-baseline + blood cultures in the previous 48 hours
- *Candida* species is known
- No mucositis of GvHD affecting the gut > Grade 3

CONCLUSION

This unique study design evaluates two treatment regimens for IC/C. The successful outcome of this trial will support a paradigm-shifting total cell wall-active strategy of echinocandin followed by oral IBX.

For additional information, contact info@scynexis.com

(For Protocol Information- ClinicalTrials.gov Identifier: NCT05178862)