SCY-247, a novel second-generation IV/oral triterpenoid antifungal, is efficacious in the neutropenic mouse model of pulmonary mucormycosis

Teclegiorgis Gebremariam [1], Yiyou Gu [1], Sondus Alkhazraji [1], Joshua Quiran[1], Laura K. Najvar [2], Nathan P. Wiederhold [2], Thomas F. Patterson [2], Scott G. Filler [1,3], David A. Angulo [4], Ashraf S. Ibrahim [1,3*]

* ibrahim@labiomed.org

[1]The Lundquist Institute at Harbor-UCLA Med Ctr., Torrance, CA; [2] University of Texas Health Sci Ctr at San Antonio, San Antonio, TX, [3]David Geffen School of Medicine at UCLA, Los Angeles, CA, [4]SCYNEXIS, Inc., Jersey City, NJ

ABSTRACT

❖ **Background:**. Invasive mucormycosis (IM) is associated with high mortality and morbidity. SCY-247 is a second-generation IV/oral triterpenoid antifungal that inhibits fungal cell wall synthesis by targeting β-(1,3)-D-glucan synthesis. We sought to evaluate the efficacy of SCY-247 monotherapy and when combined with liposomal amphotericin B (LAMB) in a neutropenic murine model of IM.

❖ **Materials/methods:** ICR mice were immunosuppressed with cyclophosphamide (200 mg/kg, IP) and cortisone acetate (500 mg/kg, SQ) on days (-2, +3 and +8), relative to intratracheal infection with *Rhizopus delemar*. Treatment with placebo (diluent control), SCY-247 (16, 32, or 48 mg/kg, PO, BID), LAMB (10 mg/kg, IV), or SCY-247 (32 mg/kg, PO, BID) + LAMB (10 mg/kg, IV) began 16 h post infection and continued for 7 days for SCY-247 and 4 days for LAMB. Survival (n=10/group) through Day +21 and tissue fungal burden (n=10/group) on Day +4 (conidial equivalents [CE]/gram of tissue using qPCR) served as primary and secondary endpoints, respectively.

❖ **Results;** SCY-247 at doses of 32 or 48 mg/kg was as effective as LAMB in prolonging median survival time (MST) and enhancing overall survival compared to placebo-treated mice (MST and survival of 8.5 Days, and 0%; 14.5 Days and 40%; 18 Days and 50%; and 15.5 Days and 40%, for placebo, SCY-247 32 mg/kg, SCY-247 48 mg/kg; and LAMB, respectively, P<0.01, Log Rank). Importantly, the combination of SCY-247+LAMB demonstrated enhanced MST and overall survival that was significantly greater compared to all monotherapies (P<0.05; MST and survival of >21 Days, and 90%). Monotherapies (only SCY-247 at 32 or 48 mg/kg BID or LAMB) also reduced the lung and brain fungal burden by ~0.5-1.0 $\log_{10}$ CE/g compared to placebo-treated mice (P<0.05, Wilcoxon Rank Sum), while the combination of SCY-247-32+LAMB lowered the fungal burden by ~1.5-2.5 $\log_{10}$ CE/g within each organ when compared to placebo or any of the monotherapy groups (P<0.007).

❖ **Conclusions:**SCY-247 demonstrated *in vivo* efficacy in treating *R. delemar* pulmonary infection in immunosuppressed mice, which was equivalent to that of antifungals currently used clinically against mucormycosis. Importantly, survival was enhanced when SCY-247 32 mg/kg was combined with LAMB 10 mg/kg. Continued investigation of SCY-247 as a novel antifungal agent against mucormycosis is warranted.

INTRODUCTION/AIMS

- Invasive mucormycosis (IM) is associated with high mortality and morbidity, and commonly afflicts patients with weakened immune systems.
- SCY-247 is a second-generation IV/oral triterpenoid antifungal that inhibits fungal cell wall synthesis by targeting β-(1,3)-D-glucan synthesis.
- Here, We sought to evaluate the efficacy of SCY-247 monotherapy and when combined with liposomal amphotericin B (LAMB) in a neutropenic murine model of IM.

METHODS

- **Immunosuppression.** Male ICR mice were immunosuppressed with cyclophosphamide (200 mg/kg, IP) and cortisone acetate (500 mg/kg, SQ) on days -2, +3 and +8, relative to infection.
- **Infection.** Immunosuppressed mice were infected with 2.5×10^5 cells of *Rhizopus delemar* by intratracheal route.
- **Treatment.** Treatment with placebo (diluent control), SCY-247 (16, 32 or 48 mg/kg, PO, BID), LAMB (10 mg/kg, IV), or SCY-247 (32 mg/kg, PO, BID) + LAMB (10 mg/kg, IV) began 16 h post infection and continued for 7 days for SCY-247 and 4 days for LAMB.
- **Efficacy endpoints.** Survival (n=10/group) through Day +21 and tissue fungal burden (n=10/group) on Day +4 (conidial equivalents [CE]/gram of tissue using qPCR) served as primary and secondary endpoints, respectively.
- **Statistical Analysis.** The nonparametric log-rank test was used to determine differences in survival times. Differences in lung, and brain CE were compared by the nonparametric Wilcoxon rank-sum test. A *P* value < 0.05 was considered significant.

REFERENCES

Ibrahim AS, *et al.* Caspofungin inhibits Rhizopus oryzae 1,3-beta-D-glucan synthase, lowers burden in brain measured by quantitative PCR, and improves survival at a low but not a high dose during murine disseminated zygomycosis. AAC 2005; 49: 721-7.

RESULTS

Figure 1. SCY-247 mono- or combination therapy improves survival of neutropenic mice with pulmonary mucormycosis. Survival of mice (n=10) infected intratracheally with *R. delemar* 99-880 (2.6 x 10^4/mouse); **Table1** median survival time, overall % survival of mice in each group. **Table 2** is the *P* values calculated by Log-rank (Mantel-Cox) test. For Figure 1 **P* <0.02 vs. placebo; ***P*<0.05 vs. placebo, and all other monotherapy treatments.

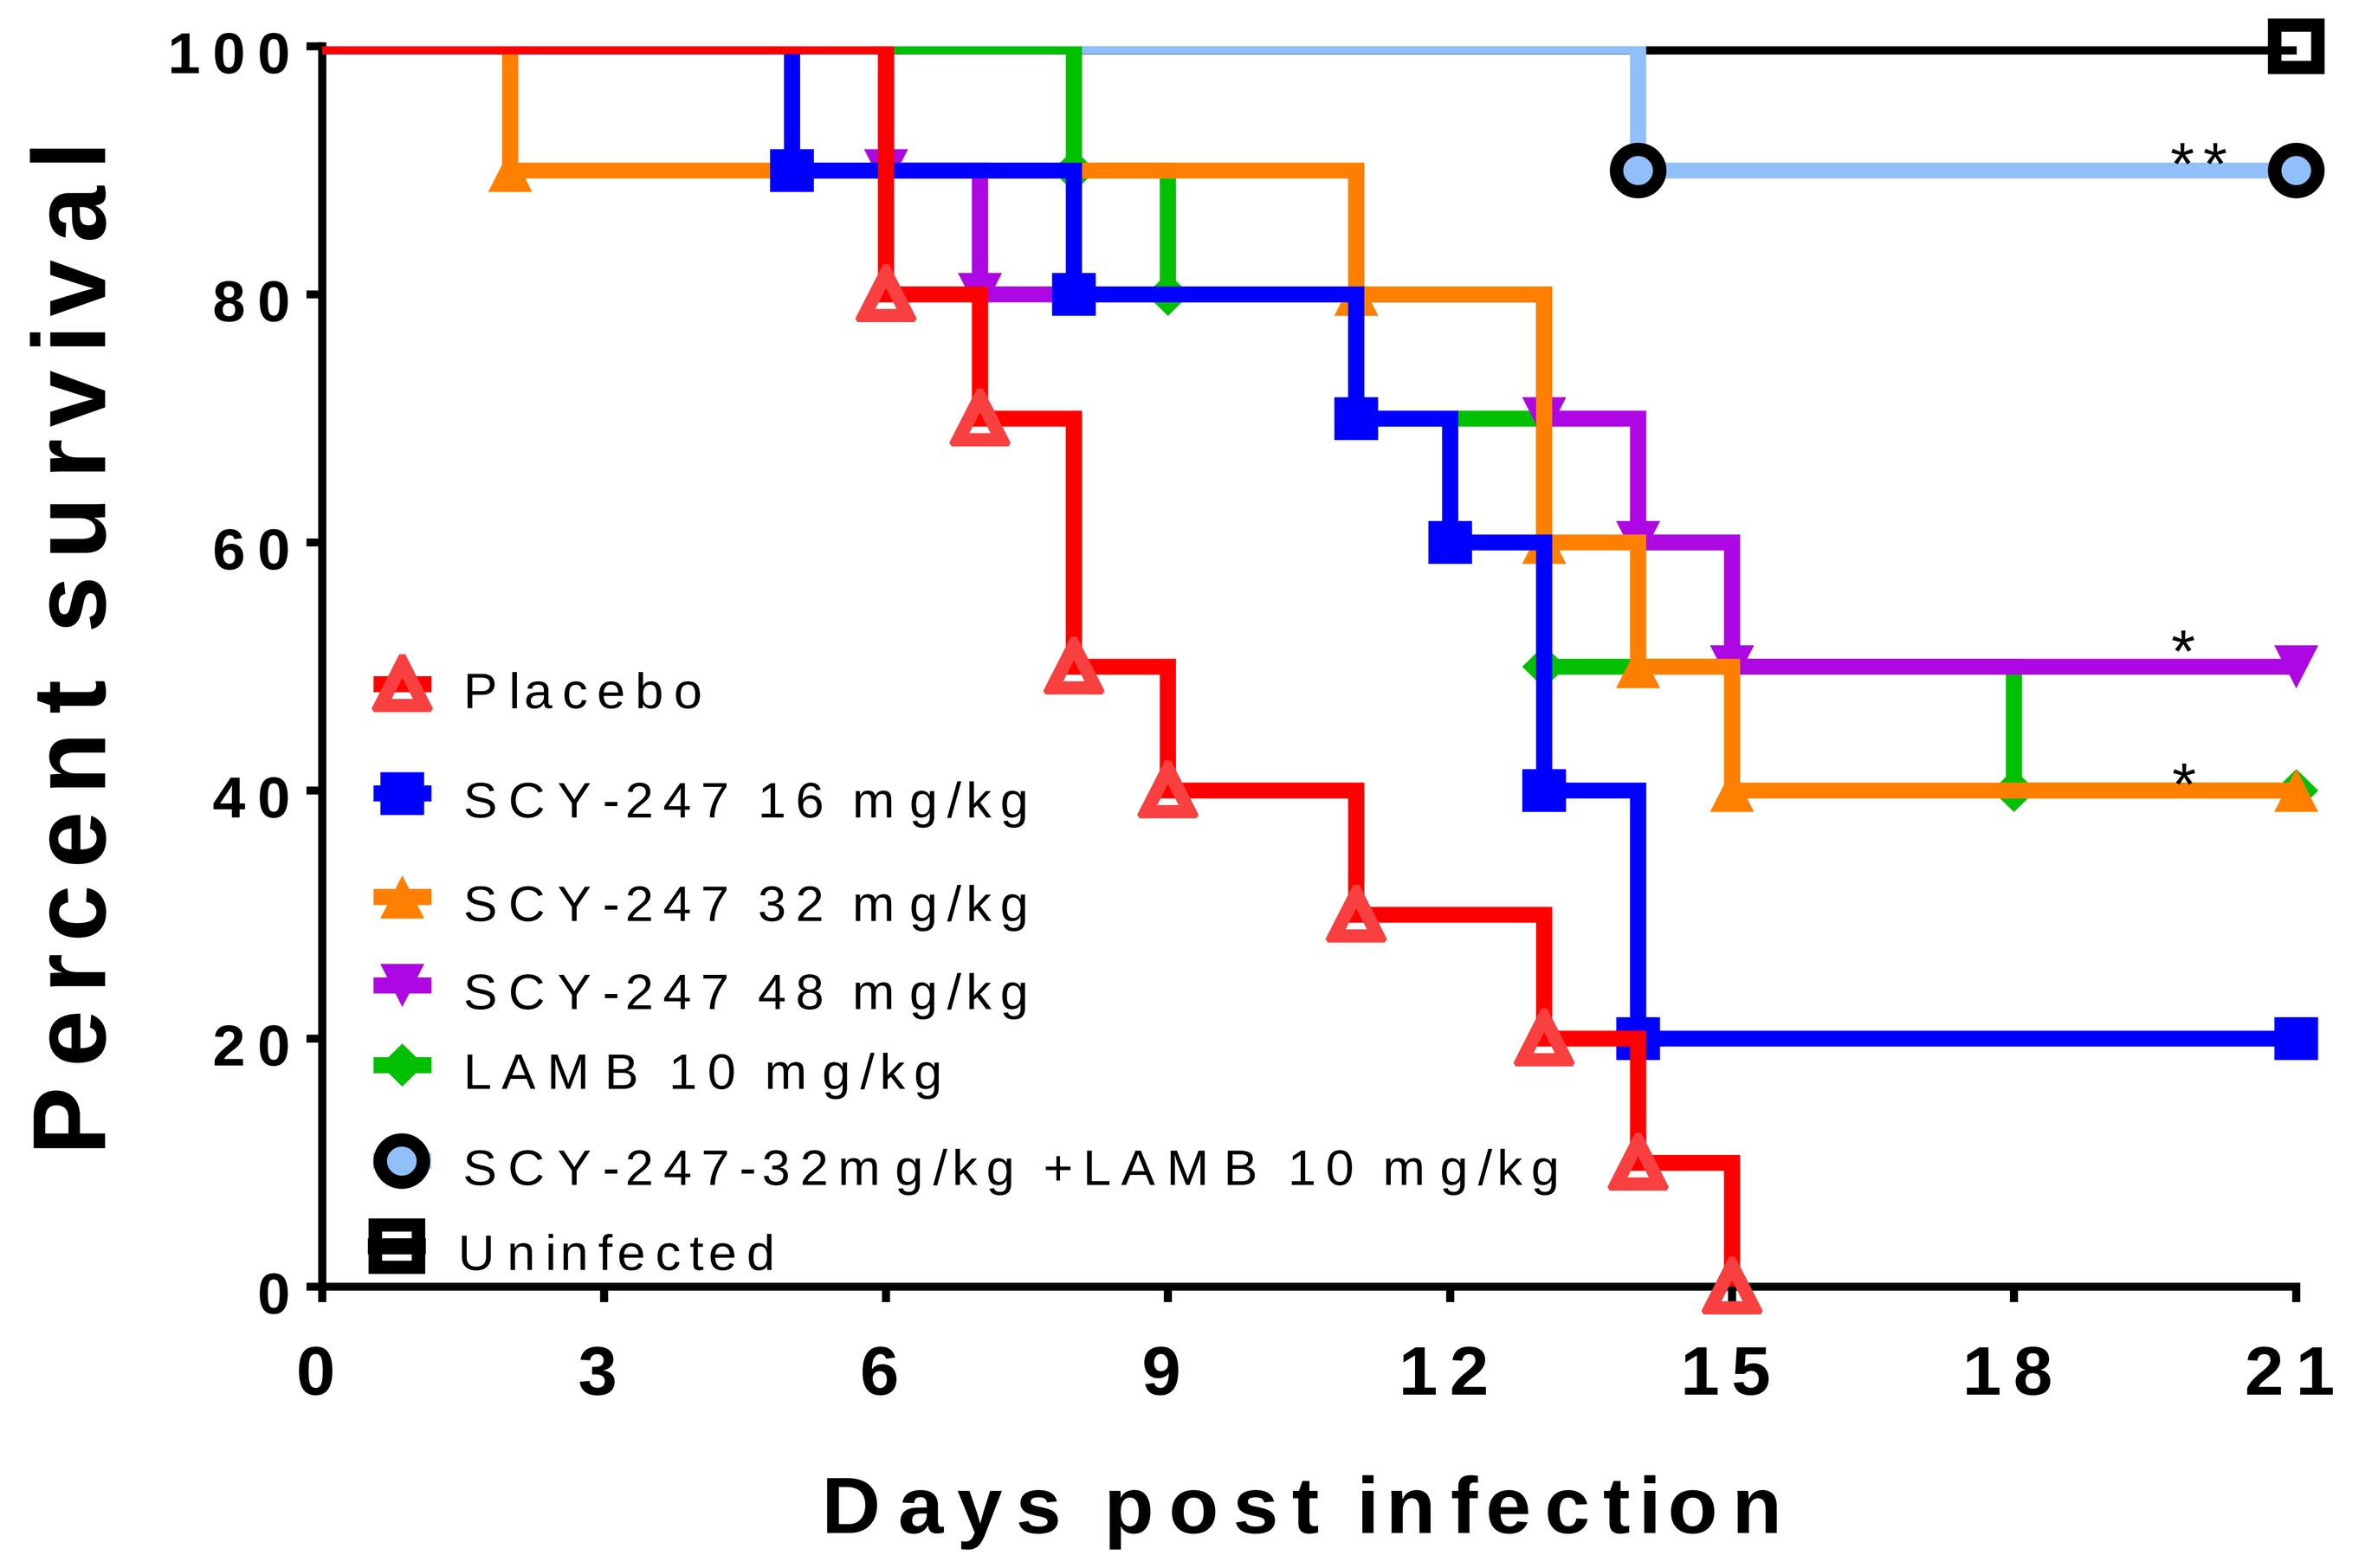

Percent survival
Days post infection
Placebo
SCY-247 16 mg/kg
SCY-247 32 mg/kg
SCY-247 48 mg/kg
LAMB 10 mg/kg
SCY-247-32mg/kg +LAMB 10 mg/kg
Uninfected

Table 1: Median Survival (Days) and % Survival

Drug dose (mg/kg)	Median survival (day)	% survival
Placebo	8.5	0
SCY-247 16mg/kg	13	20
SCY-247 32mg/kg	14.5	40
SCY-247 48mg/kg	18	50
L-AMB 10mg/kg	15.5	40
SCY-247-32 + LAMB 10mg/kg	>21	90

Table 2: Log-rank (Mantel-Cox) test

	Placebo	SCY-247 16mg/kg	SCY-247 32mg/kg	SCY-247 48mg/kg	LAMB 10mg/kg	SCY-247 32+ LAMB
Placebo		0.115	0.011	0.007	0.008	< 0.0001
SCY-247 16mg/kg	0.115		0.280	0.155	0.373	0.001
SCY-247 32mg/kg	0.011	0.280		0.677	0.925	0.018
SCY-247 48mg/kg	0.007	0.155	0.677		0.703	0.049
LAMB 10mg/kg	0.008	0.373	0.925	0.703		0.017
SCY-247 32+ LAMB	< 0.0001	0.001	0.018	0.049	0.017	

Figure 2. SCY-247 mono (32 or 48 mg/kg) or combination therapy reduced tissue fungal burden of immunosuppressed mice infected with *R. delemar*. Tissue fungal burden of lung and brain of mice infected intratracheally with *R. delemar* 99-880 (4.1 x 10^3/mouse). Mice were sacrificed on Day +4, relative to infection. Statistical analysis was conduced with Wilcoxon Rank Sum test**.**

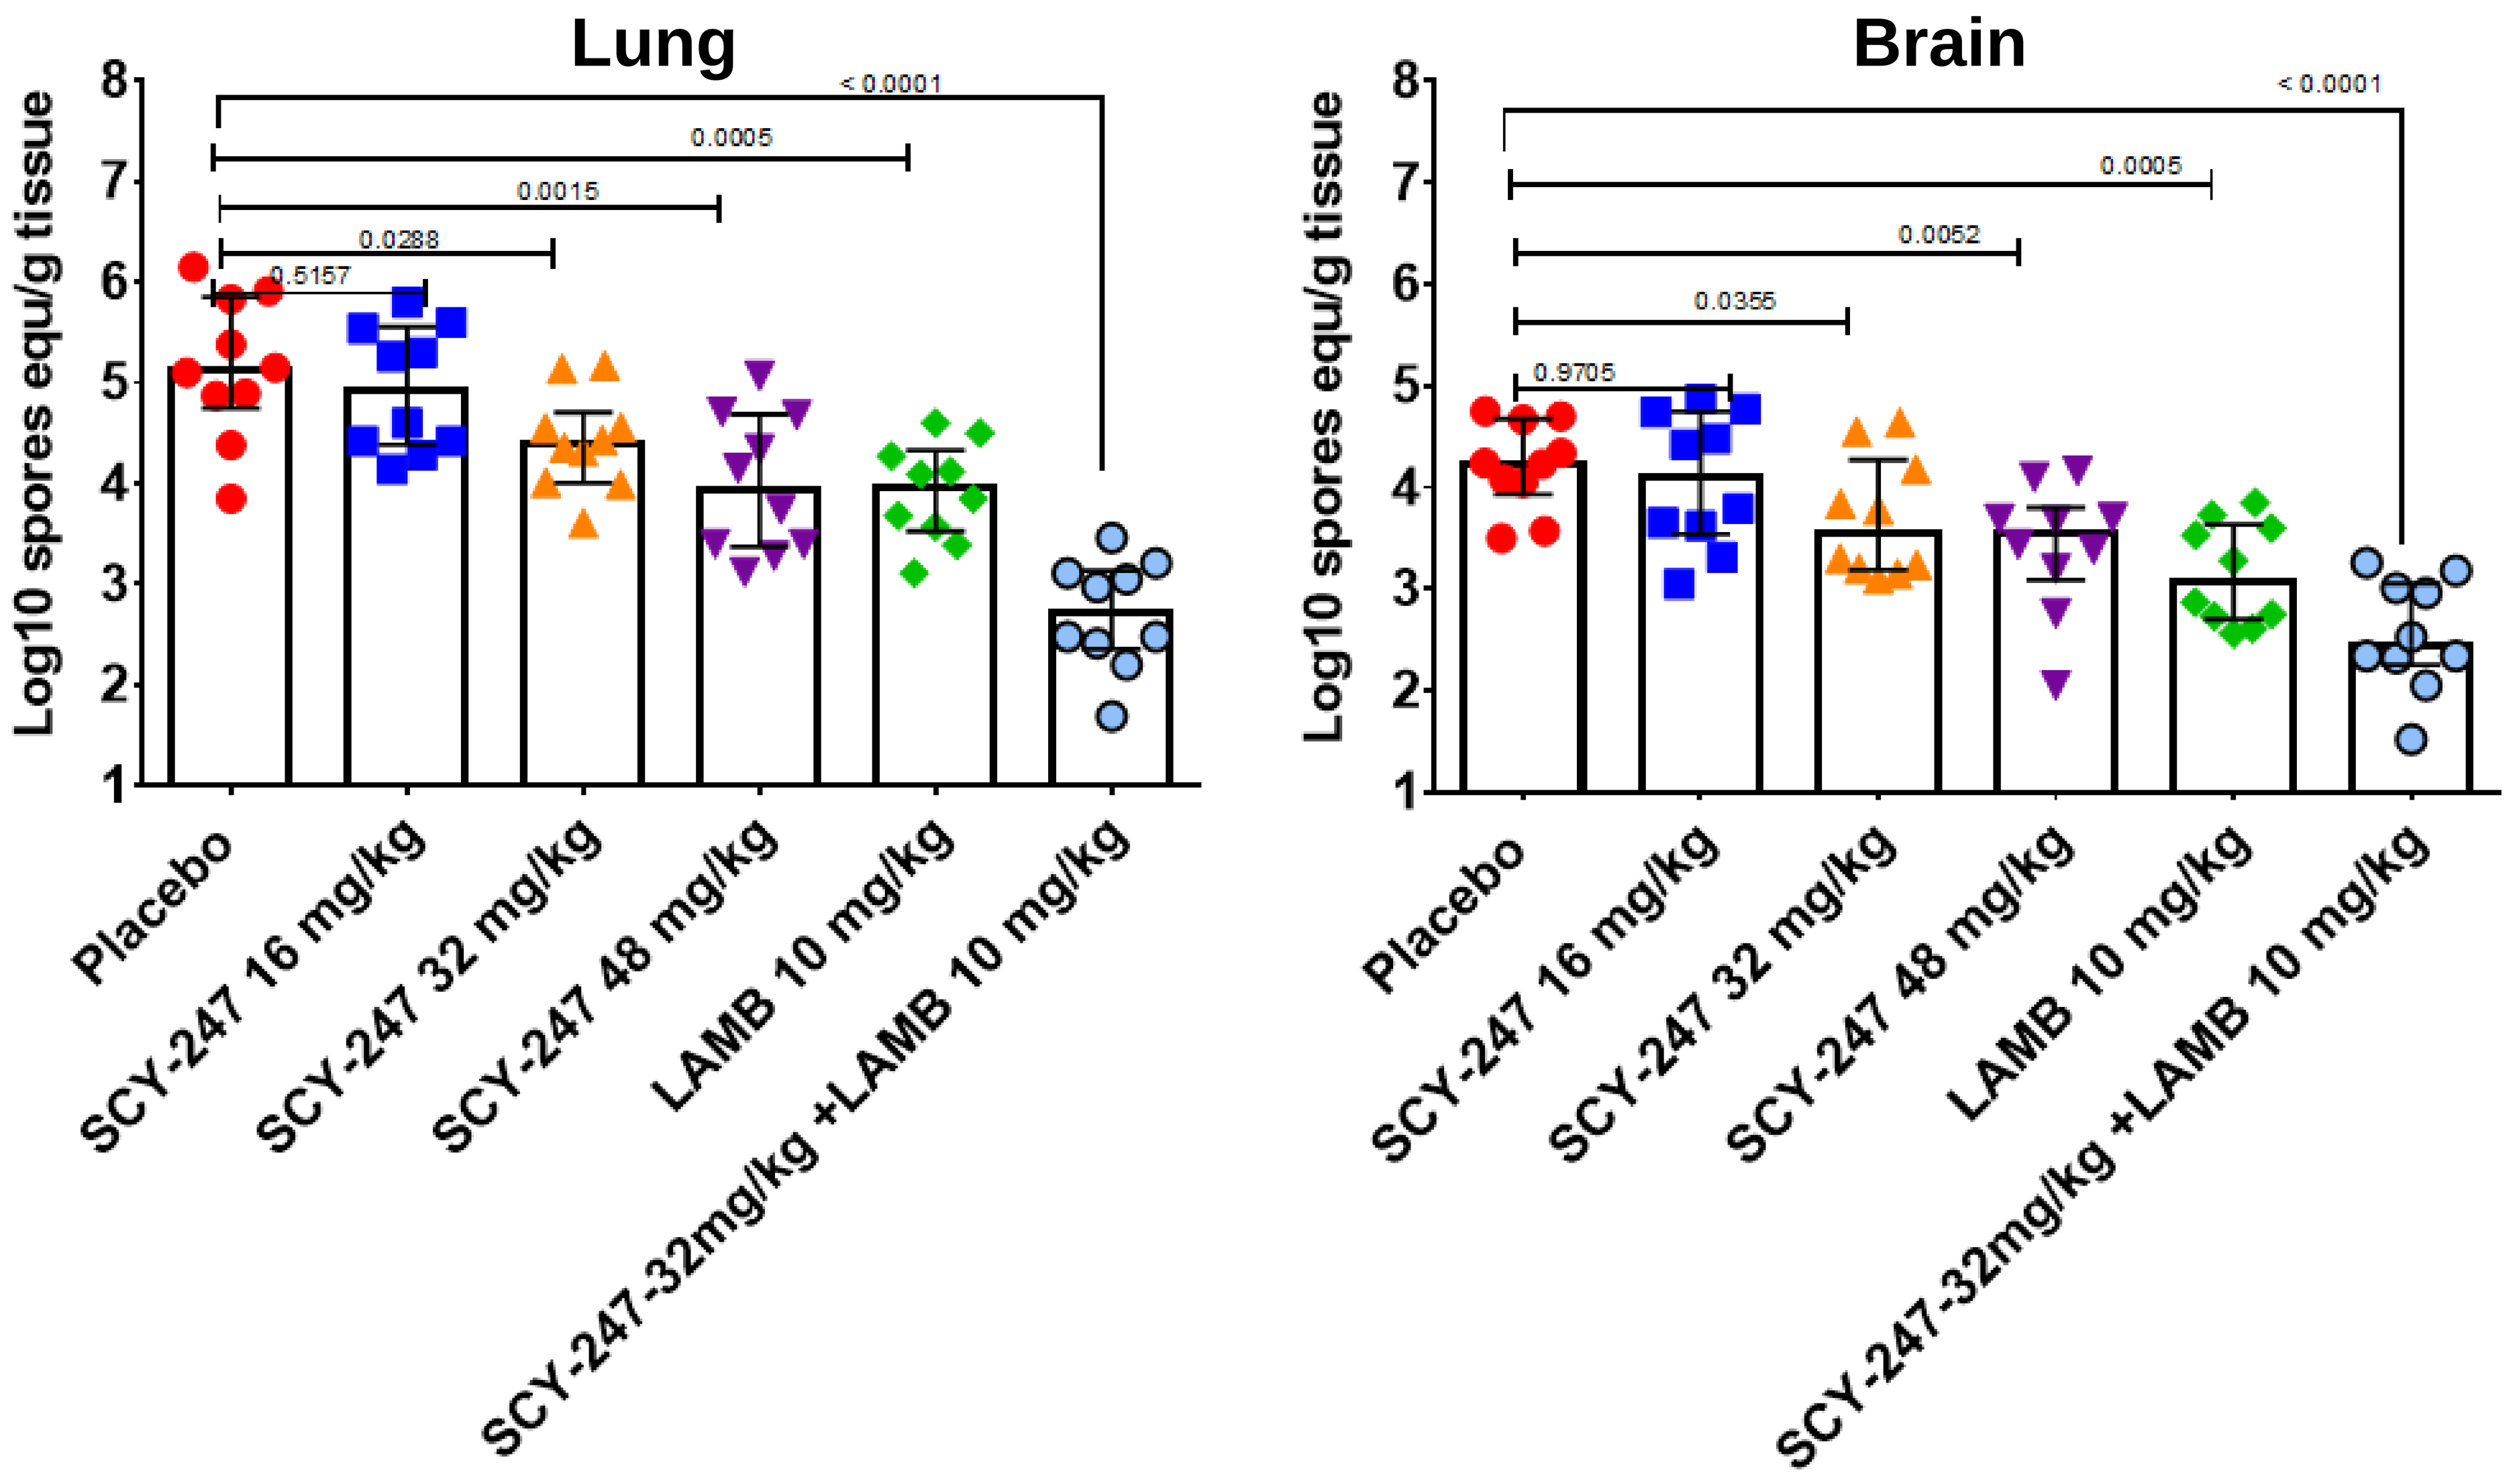

Lung
Brain
Log10 spores equ/g tissue
< 0.0001
0.0005
0.0015
0.0288
0.5157
0.0052
0.0355
0.9705
Placebo
SCY-247 16 mg/kg
SCY-247 32 mg/kg
SCY-247 48 mg/kg
LAMB 10 mg/kg
SCY-247-32mg/kg +LAMB 10 mg/kg

SUMMARY/CONCLUSIONS

- SCY-247 demonstrated *in vivo* efficacy in treating *R. delemar* pulmonary infection in immunosuppressed mice, which was equivalent to that of antifungals currently used in clinical management of mucormycosis.
- Importantly, SCY-247 demonstrated synergy when combined with LAMB against murine mucormycosis, caused by *R. delemar*.
- Continued investigation of SCY-247 as a novel antifungal agent against mucormycosis is warranted.

ACKNOWLEDGEMENT

This research support was provided by the NIH/NIAID under Contract No. HHSN272201700039I (Task order A65 75N93022F00001) and by NIAID R01 AI063503.